High Resolution Orbitrap Mass Spectrometry: Method Development and Qualification of a Synthetic Oligoribonucleotide

Robert J. Duff, Ph.D. and Jon S. Kauffman, Ph.D.
Lancaster Laboratories Biochemistry Department

Background on LTQ-Orbitrap

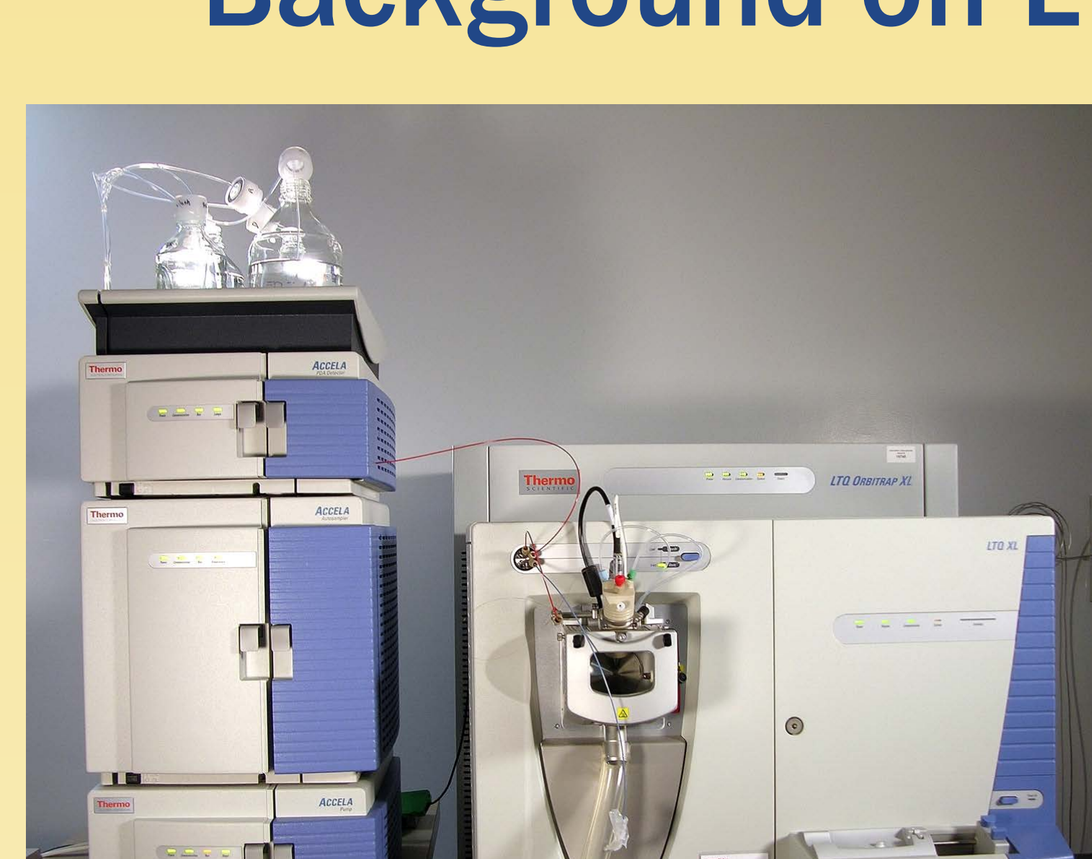

- Orbitrap: an electrostatic ion trap (magnet-free Fourier Transform Mass Spectrometry)
 - First new type of mass spectrometer in more than 20 years
- Two Detectors in One Instrument
 - Front End - LTQ has all the features of a linear trap
 - Back End - Orbitrap - High Mass Resolution (>100,000) and Sub-ppm mass accuracy with external calibration
- LTQ-Orbitrap Performance
 - Mass resolution: >60,000 at m/z 400 at 1 sec
 - <5 ppm external calibration
 - Throughput: 3 High resolution scans/second
- Accela is an U-HPLC
 - High Pressure Mode
 - 7000-16000 psi
 - Low flow (25 µL/min)

Nucleic Acid Chemistry

- Synthesis of Oligoribonucleotide - Thermal Deprotection Step
 - Cieslak et al. 2008
 - 4-(N-Dichloroacetyl-N-methylamino)benzyloxymethyl group

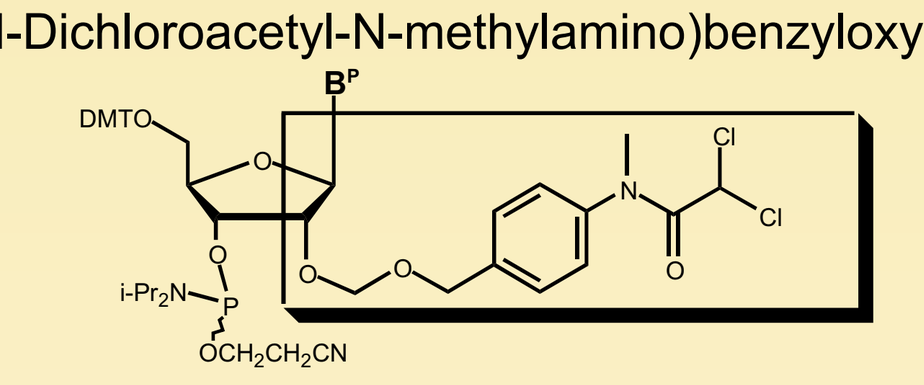

DMTO
B^P
i-Pr_2N
OCH_2CH_2CN

- RNA Oligonucleotide – 20-mer - Ave Mass: 6366.863g/mole
 - AUCCGUAGCUAACGUCAUGG
 - Commercial Source
 - Beaucage Oligoribonucleotide/Synthesis
 - N-1, N-2 - 5' Failure Sequences
 - UCCGUAGCUAACGUCAUGG
 - CCGUAGCUAACGUCAUGG

OBJECTIVE

– To develop and qualify a high mass resolution LC/MS/MS method for the characterization of a RNA oligonucleotide (5'AUCCGUAGCUAACGUCAUGG) in order to compare the quality of a commercial source with the same oligoribonucleotide produced through the use of the novel 2'-hydroxyl group, 4-(N-dichloroacetyl-N methylamino)benzyloxymethyl group

METHODOLOGY

This method utilized a hybrid linear ion trap, the Thermo Scientific LTQ XL™ Orbitrap mass spectrometer (LC-ESIMS/MS) as used in conjunction with an Accela™ high-speed chromatographic system. Development began with U-HPLC separation using reverse phase C18 columns (Phenomenex Luna (250 x 2.0 mm, flow 0.2 mL/min) and Supelcosil LC-18S column (250 x 4.6 mm, flow 0.8 mL/min)). The oligoribonucleotides were eluted using various mobile phases and conditions. Two mobile phase compositions were primarily examined: 1) triethylammonium bicarbonate (TEAB), pH 7.0 in acetonitrile and 2) 1,1,1,3,3,3-hexafluoro-2-propanol/triethylamine (pH 7.0) in methanol. Sequence analysis was carried out using through MSn analyses number. ESI mass spectra were acquired in negative ion mode.

- LTQ-Orbitrap with Accela
 - Direct Infusion Conditions
 - RNA Solution – 4 µM Oligonucleotide in 5% Triethylamine/25%CH_3CN/70% RNase-free Water
 - Infused at 10 µL/min
 - LC flow 200 µL/min, 50% CH_3OH/Water with 100 mM HFIP/8.6 mM TEA added
 - Tuned for maximum LC/MS sensitivity
 - Mass Spectrometer Parameters
 - Electrospray Voltage: 4 kV
 - Capillary Voltage: –67.35 V
 - Sheath Gas: 60 arbitrary units
 - Aux N_2 Gas: 0 arbitrary units
 - Capillary Temp: 200 °C
 - Xcaliber 2.0.7 on Windows XP Platform
 - ProMass 2.5 SR-1
 - Accela Parameters
 - High Pressure Mode (7000 psi to 16000 psi)
 - Column Compartment: Ambient
 - Autosampler Temperature: 5 °C
 - Flow Monitored 260 nm

RESULTS

Observed Ion Analysis

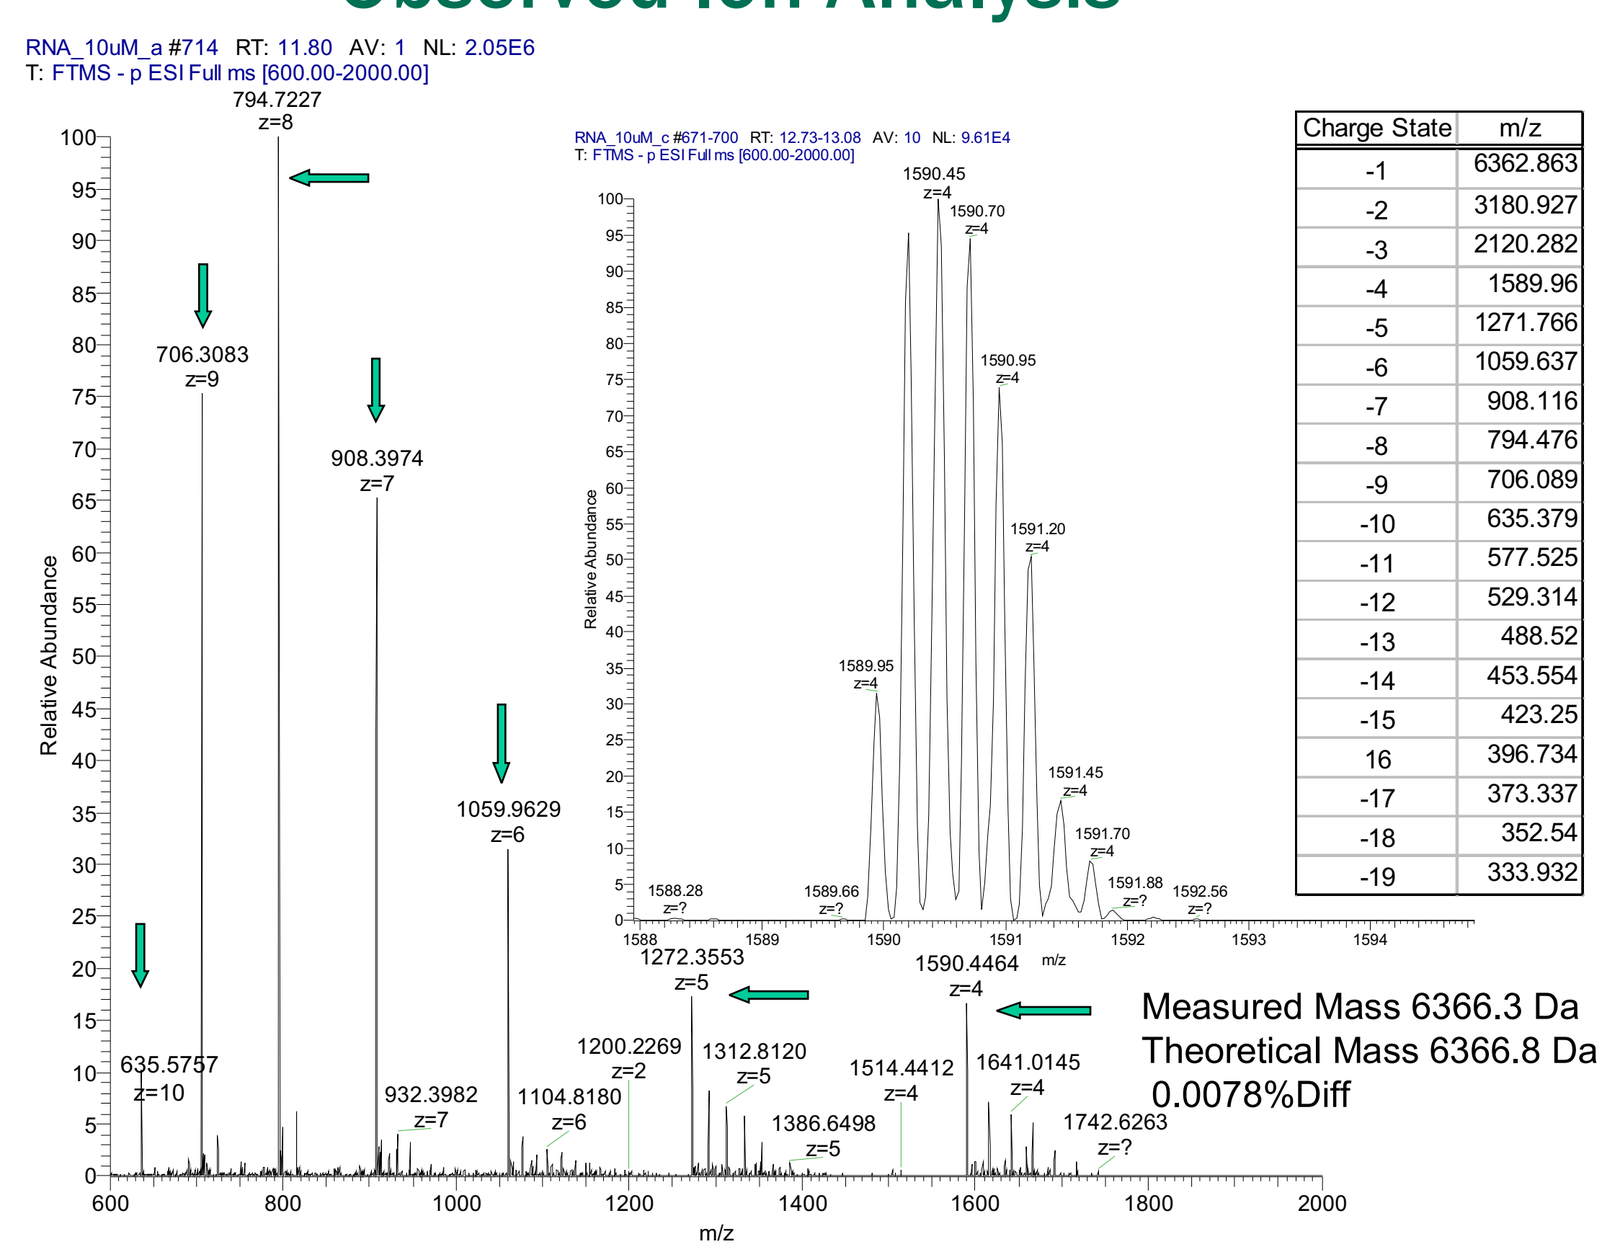

RNA_10uM_a #714 RT: 11.80 AV: 1 NL: 2.05E6
T: FTMS - p ESI Full ms [600.00-2000.00]
794.7227 z=8
706.3083 z=9
908.3974 z=7
1059.9629 z=6
1272.3553 z=5
1590.4464 z=4
Relative Abundance
m/z

Charge State	m/z
-1	6362.863
-2	3180.927
-3	2120.282
-4	1589.96
-5	1271.766
-6	1059.637
-7	908.116
-8	794.476
-9	706.089
-10	635.379
-11	577.525
-12	529.314
-13	488.52
-14	453.554
-15	423.25
16	396.734
-17	373.337
-18	352.54
-19	333.932

Measured Mass 6366.3 Da
Theoretical Mass 6366.8 Da
0.0078%Diff

U-HPLC Analysis of RNA

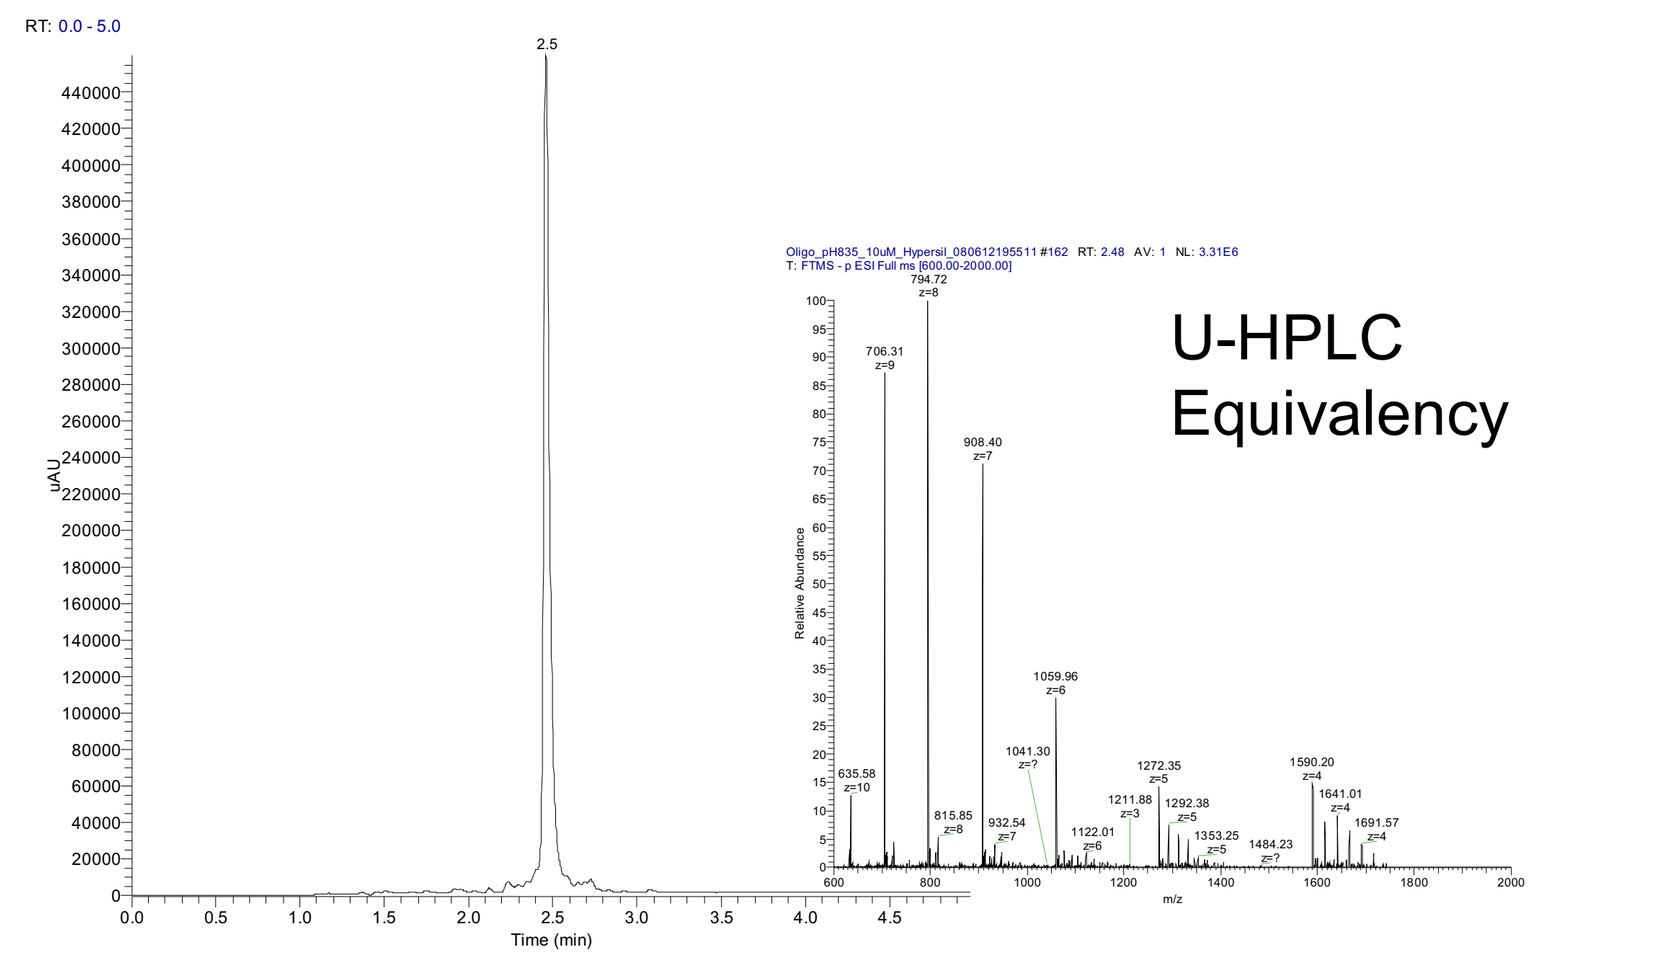

U-HPLC Equivalency
Time (min)

Full scale (Left) and Mass chromatogram (Inset) of RNA 20-mer. Conditions: Sample Load: 5 µL, Flow rate: 477 µL/min, Temperature: 25ºC, Mobile Phase A Composition: 100 mM hexafluoroisopropanol (HFIP)/ triethylamine (TEA) **(pH 8.35)** in RNase-free MQ H_2O; Mobile Phase B Composition 100 mM hexafluoroisopropanol (HFIP)/ triethylamine (TEA) in CH_3OH. Starting Composition: 98%A, Elution Gradient: 19.6% B / min (2% B to 100% B in 5 min), Monitored 260 nm , Thermo Hypersil GOLD Column (50 mm x 2.1 mm), 1.9 micron particle size.

Sequence Analysis

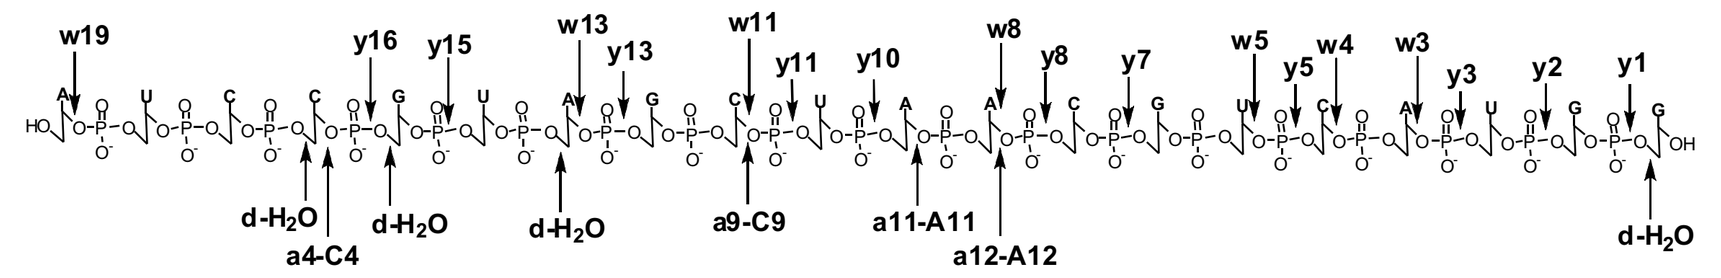

w19 y16 y15 w13 y13 w11 y11 y10 w8 y8 y7 w5 y5 w4 w3 y3 y2 y1
d-H2O a4-C4 d-H2O d-H2O a9-C9 a11-A11 a12-A12 d-H2O

Fragment	m/z	Fragment	m/z
w3	337.0343	a11-A11	823.0974
y3	466.07	a12-A12	724.0859
M-A	777.8407	y15	794.5934
d-H_2O (n=19)	867.77	y13	826.9063
d-H_2O (n=4)	621.5697	d-H_2O (n=6)	947.101
d-H_2O (n=3)	939.1022	w8	650.075
w11	712.685	M-AH	777.9674
M-A	691.4152	y2	627.1301
a9-C9 (n=9)	893.78	y1	282.0827
a4-C4 (n=4)	1053.14	y5	783.1191
w4	670.5808	y7	738.7647
w13	702.0791	w19	763.8365
w5	548.3995	y10	794.86
y14	371.2736	y16	730.6646
y11	871.36	y8	840.7824

N-1 and N-2 Failure Sequences

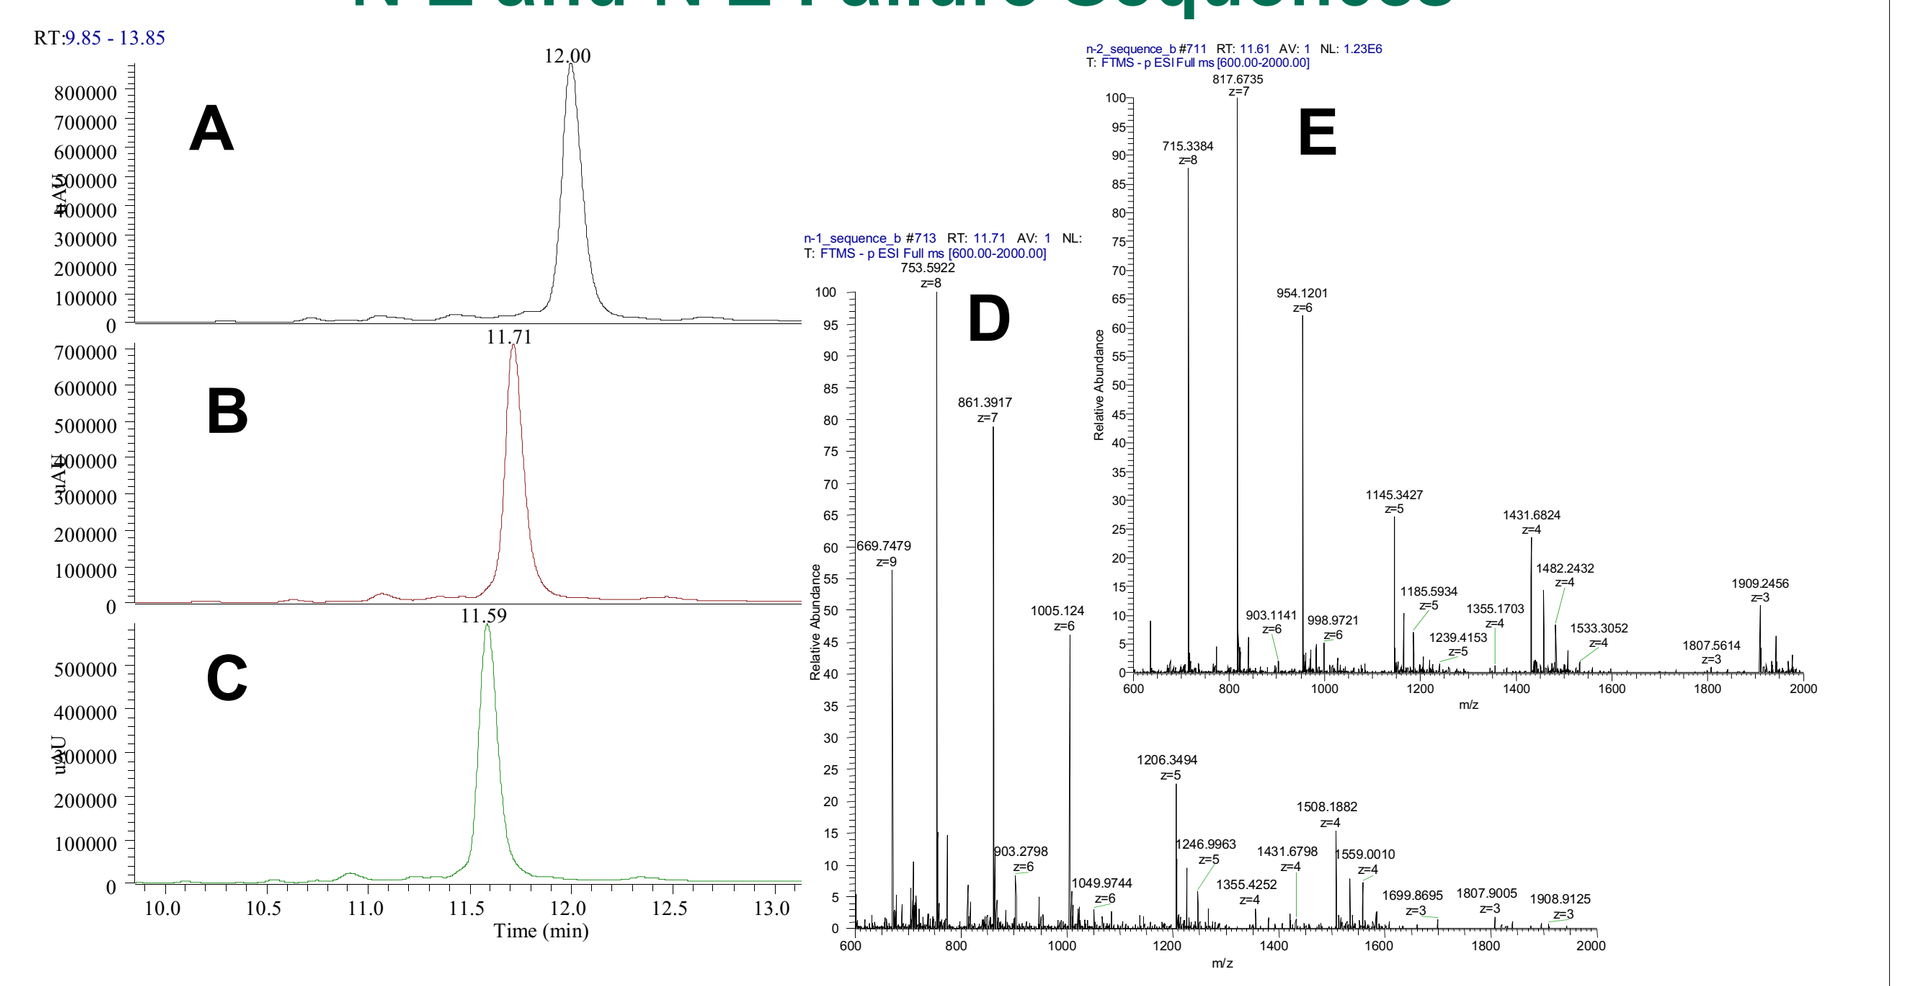

A
B
C
D
E
Time (min)

A) Full Length RNA 20 mer, B) Authentic N-1 Sequence, C) Authentic N-2 Sequence D) Full MS (600.000-2000.000) of N-1 Sequence, E) Full MS (600.000-2000.000) of N-2 Sequence

2'-Hydroxyl Protection Strategies

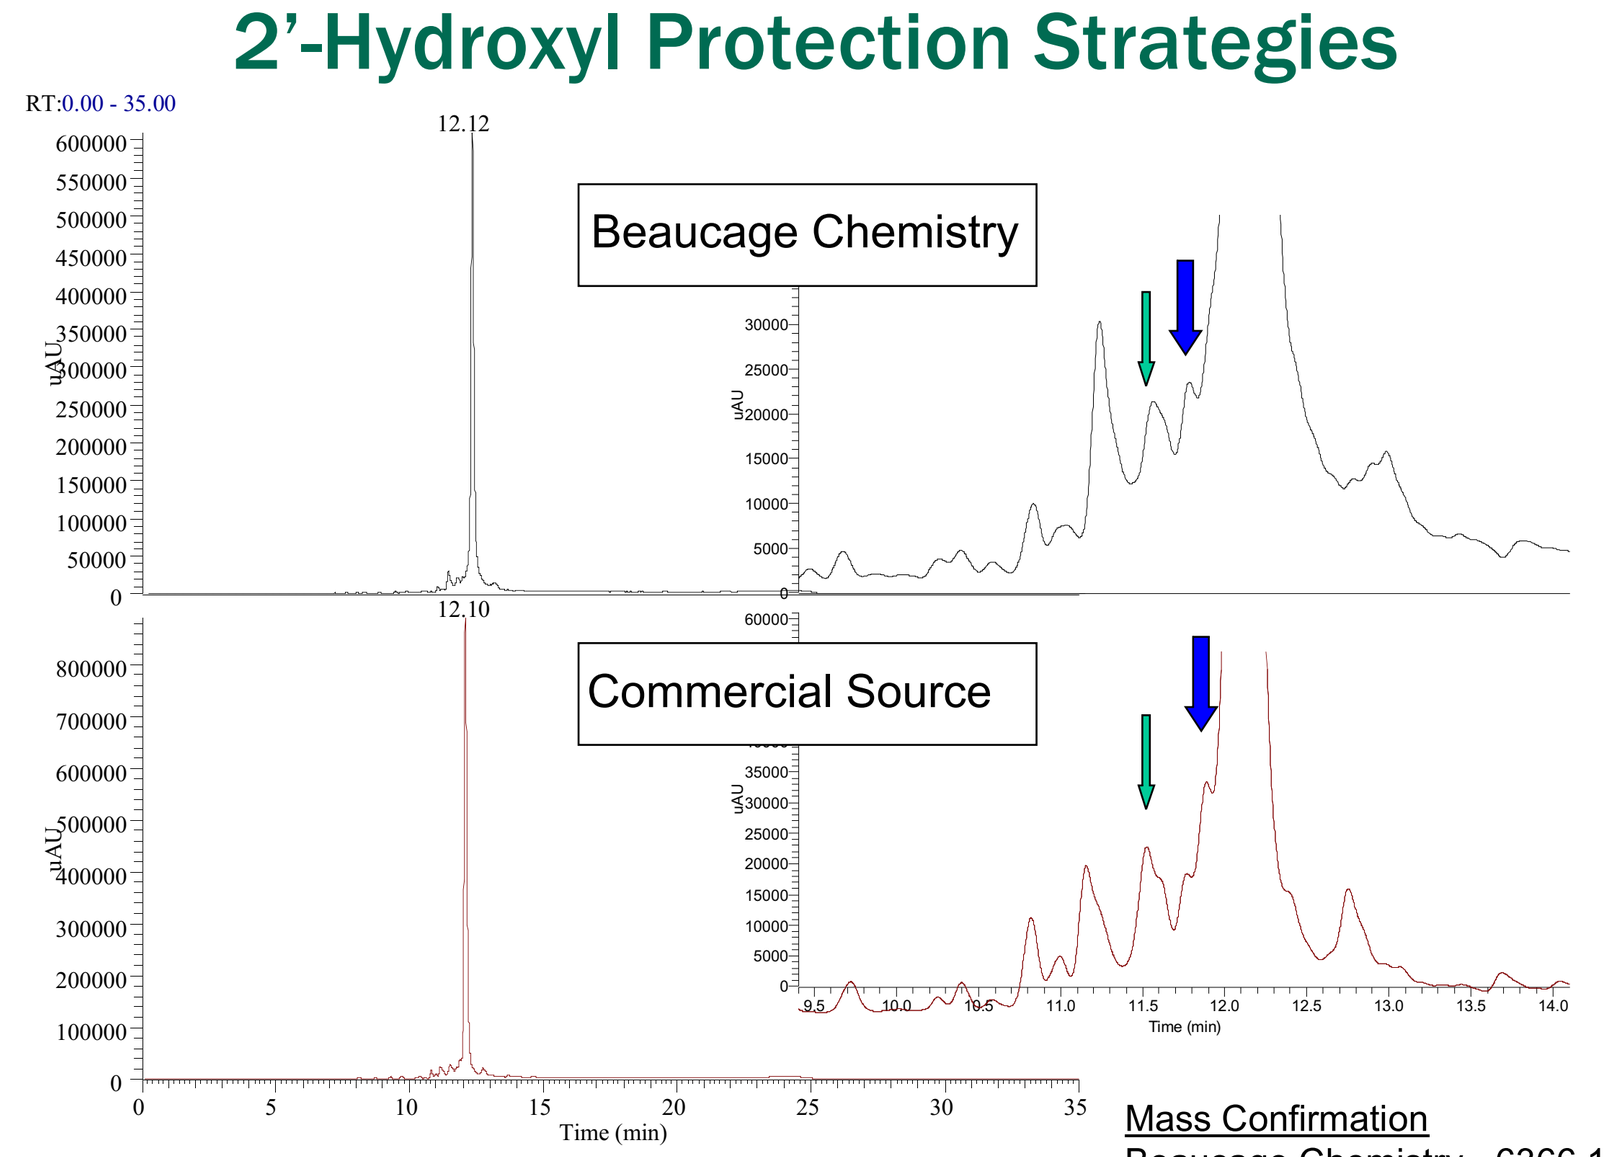

Beaucage Chemistry
Commercial Source
Time (min)

Arrows denote N-1 (blue) and N-2 (green) failure sequences

Mass Confirmation
Beaucage Chemistry - 6366.1 Da
Commercial Source - 6366.1 Da

Initial HPLC Development

- Supelcosil LC-18 (250 mm x 4.6 mm, 5 micron)
- Triethylammonium Bicarbonate Buffer (TEAB)
 - Prepared 14 mL TEA in 1800 mL of RNase-free H_2O, pH adjusted with dry ice.
 - CH_3CN
- pH 7.0
- Monitored 260 nm
- Agilent G1969 ESI-TOF

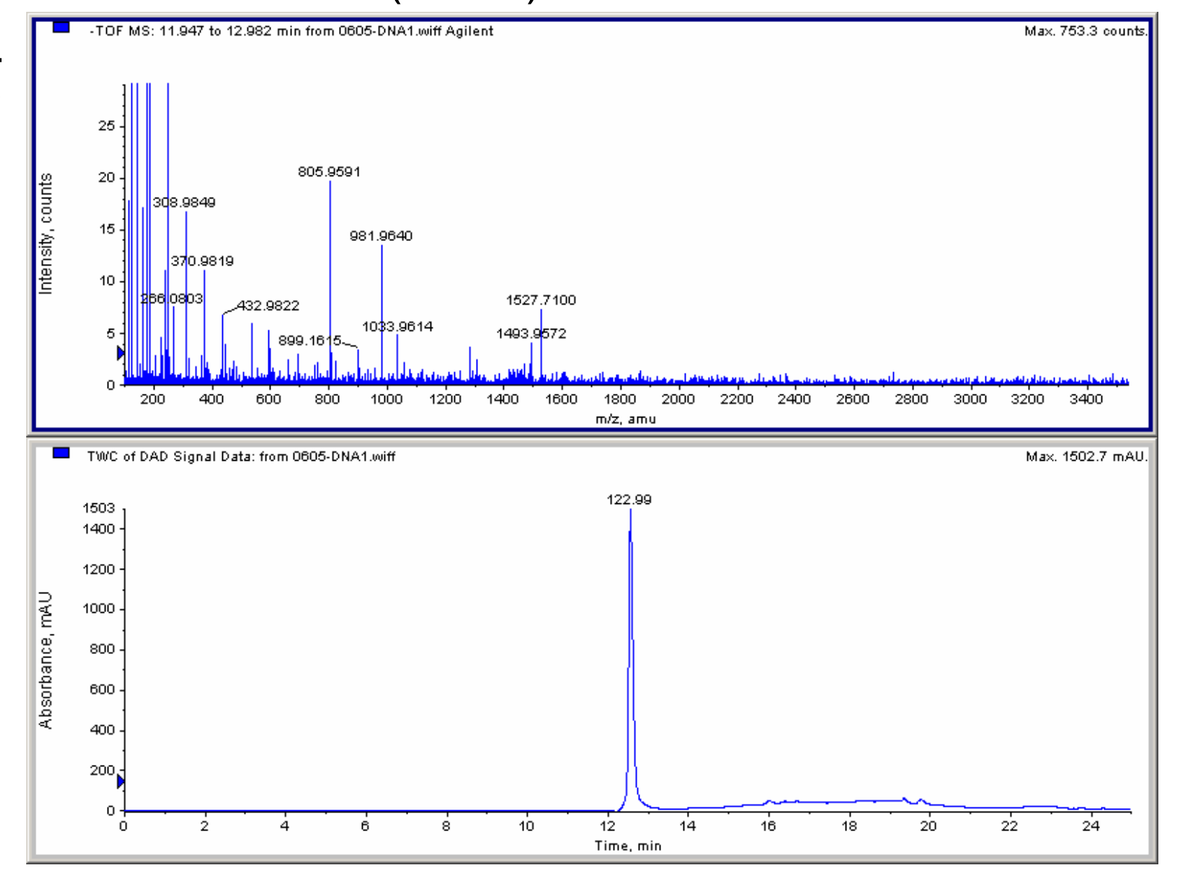

Problem:
Ion Suppression

Linearity and Accuracy

- Solution Preparation: 1 mM solution -diluted with RNase-free H_2O to 10 µM solution
 - Range 10 nM to 10 µM. 10, 25, 50, 100, 500, 1000 and 10000 nM

Linearity

- Linear over 3 orders of magnitude
- nanomolar ranges (picomoles-on-column)

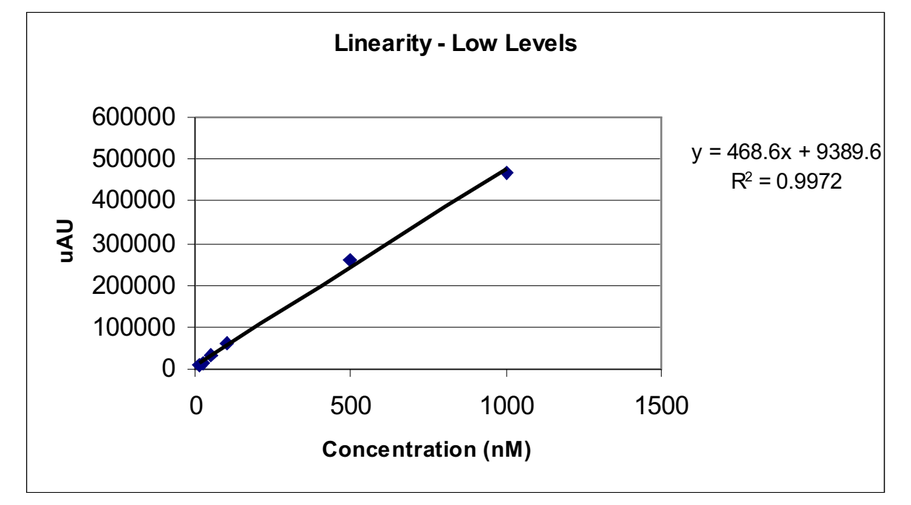

Linearity - Low Levels
y = 468.6x + 9389.6
R^2 = 0.9972
uAU
Concentration (nM)

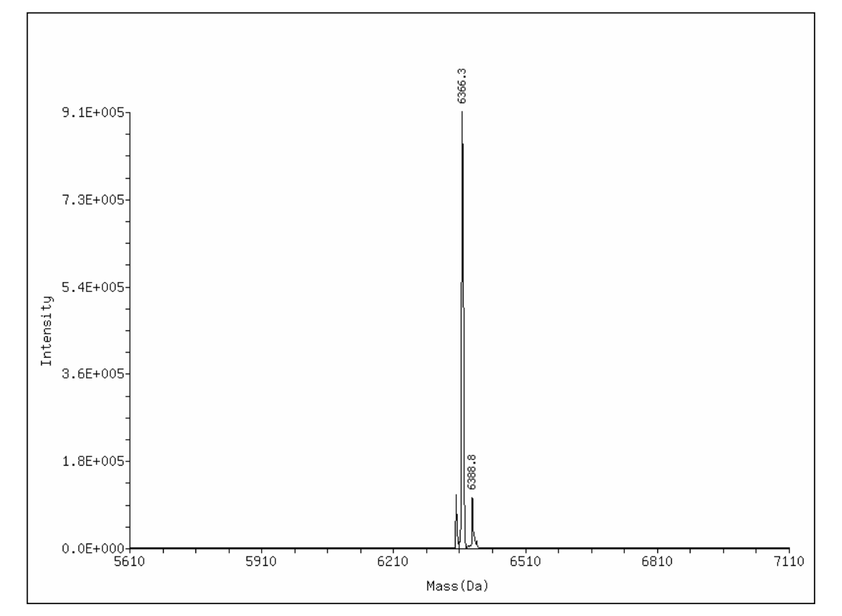

Accuracy

% Accuracy less than 0.02% of Theoretical Mass

Run Number	Average Mass (Da)
RNA Run #1	6366.000
RNA Run #2	6366.000
RNA Run #3	6366.000
RNA Run #4	6366.100
RNA Run #5	6366.000
RNA Run #6	6366.200
Average (Da)	6366.050
St. Dev	0.084
%RSD	0.001
Observed (Da)	6366.05
Theoretical (Da)	6366.889
Accuracy%	0.013

MSⁿ: MS/MS of -9 Charge State

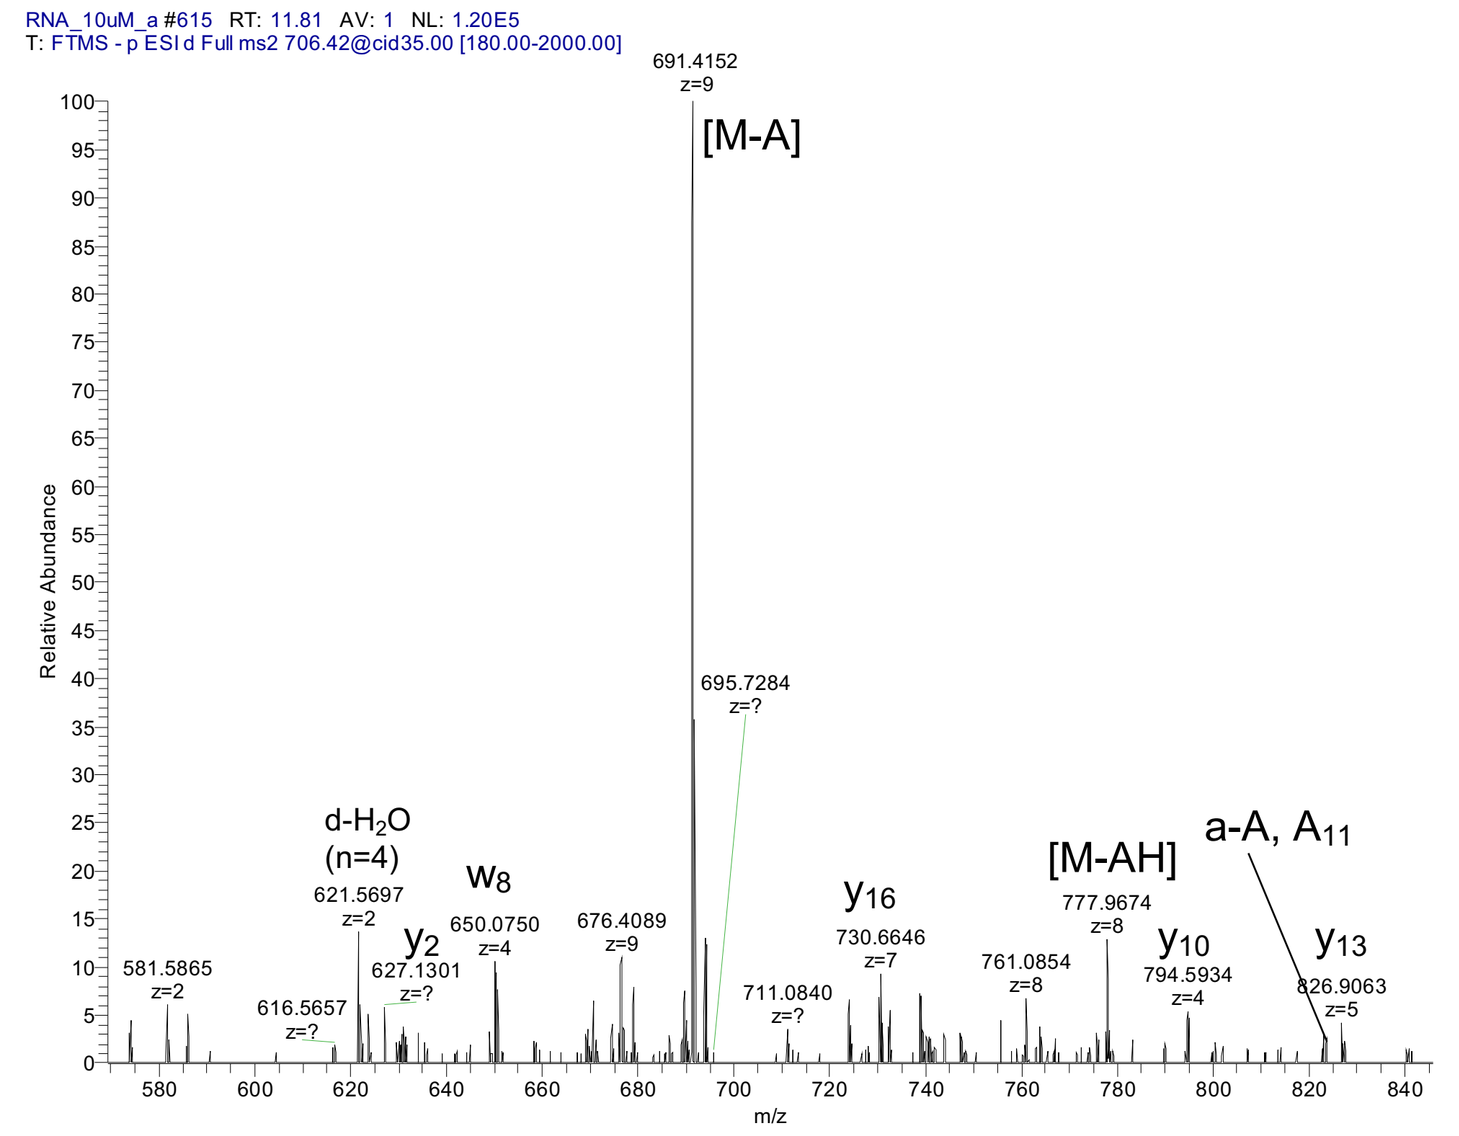

RNA_10uM_a #615 RT: 11.81 AV: 1 NL: 1.20E5
T: FTMS - p ESI d Full ms2 706.42@cid35.00 [180.00-2000.00]
691.4152 z=9
[M-A]
d-H2O (n=4)
w8
y2
y16
[M-AH]
a-A, A11
y10
y13
Relative Abundance
m/z

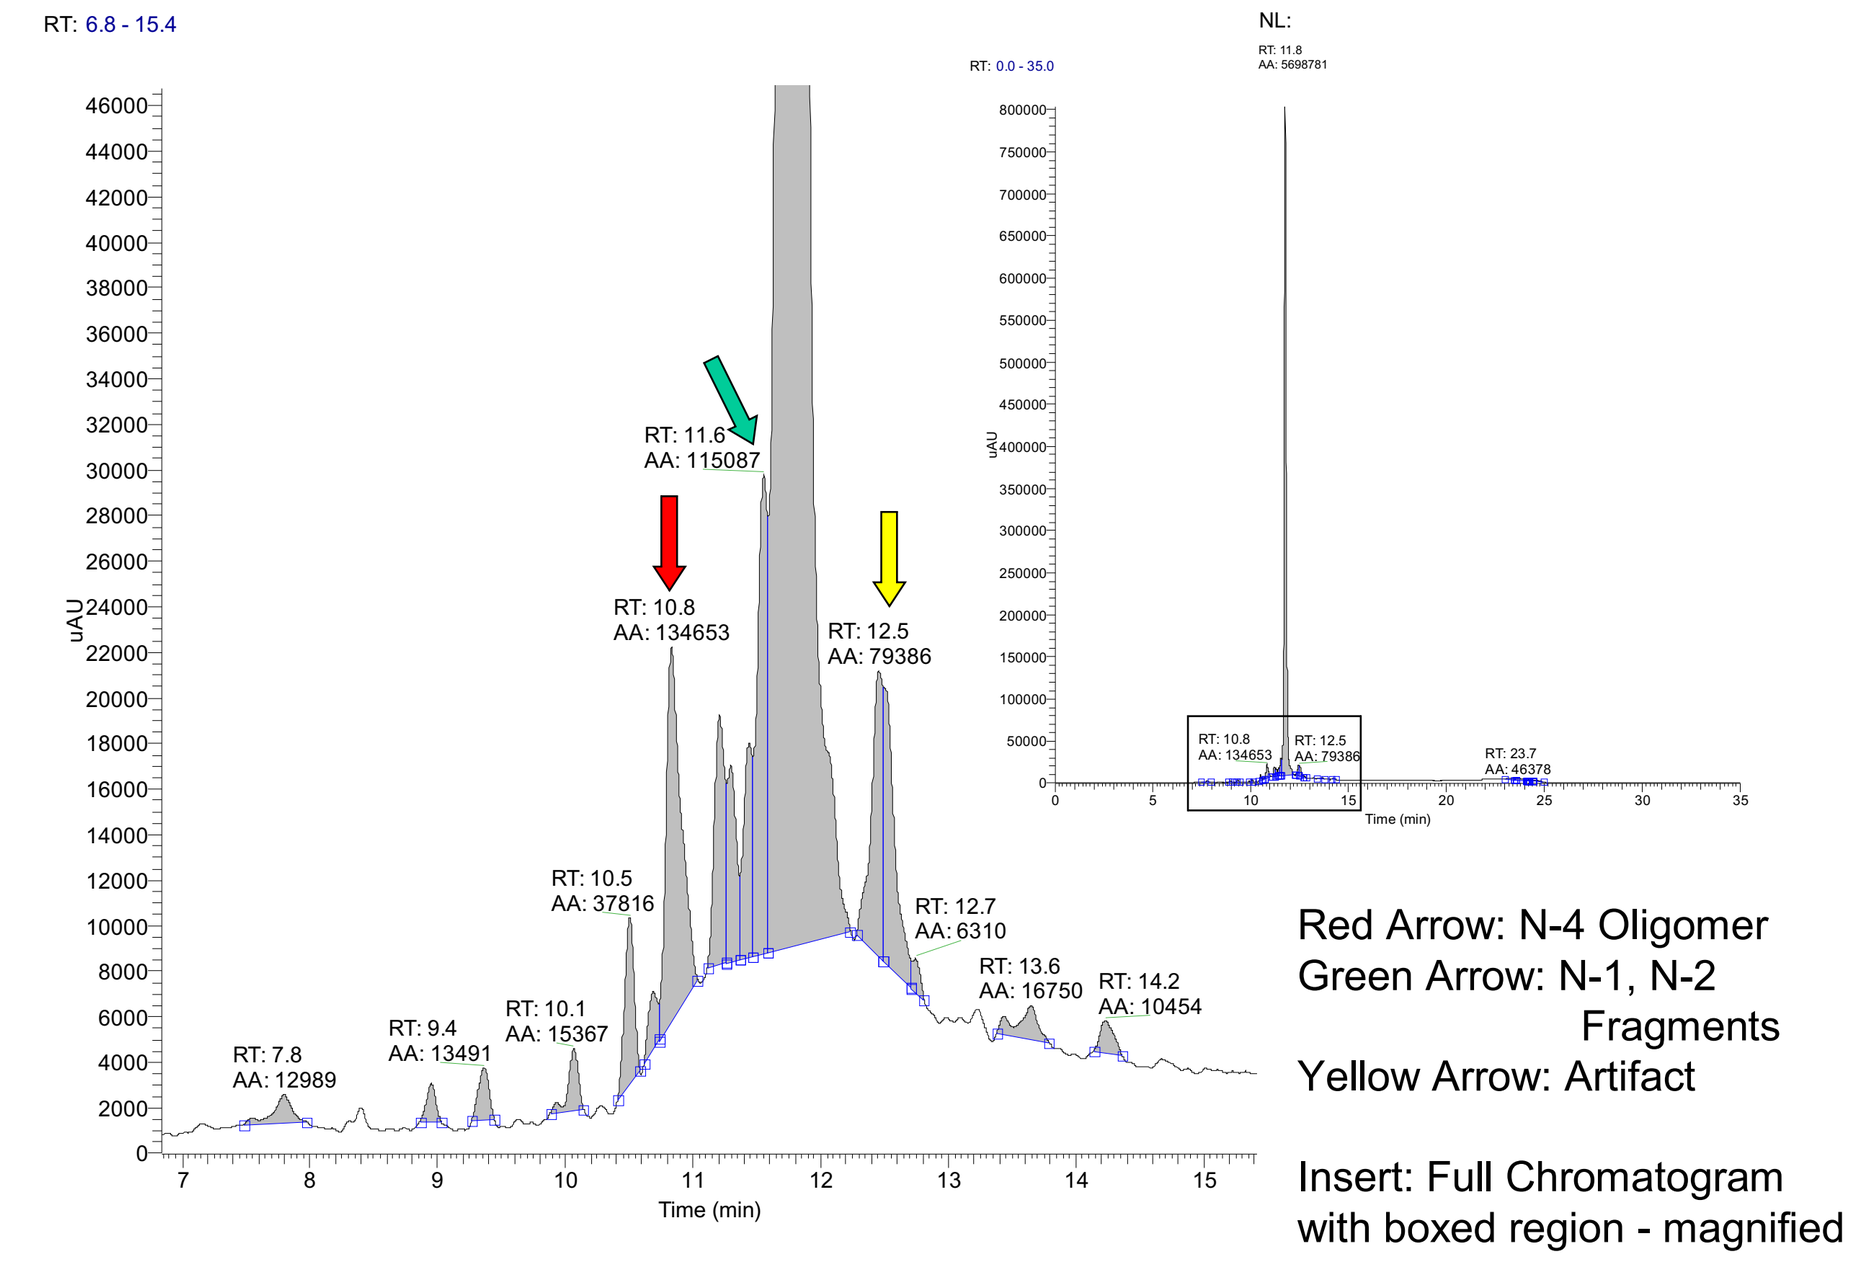

RT: 10.8 AA: 134653
RT: 11.6 AA: 115087
RT: 12.5 AA: 79386
Time (min)

Red Arrow: N-4 Oligomer
Green Arrow: N-1, N-2 Fragments
Yellow Arrow: Artifact

Insert: Full Chromatogram with boxed region - magnified

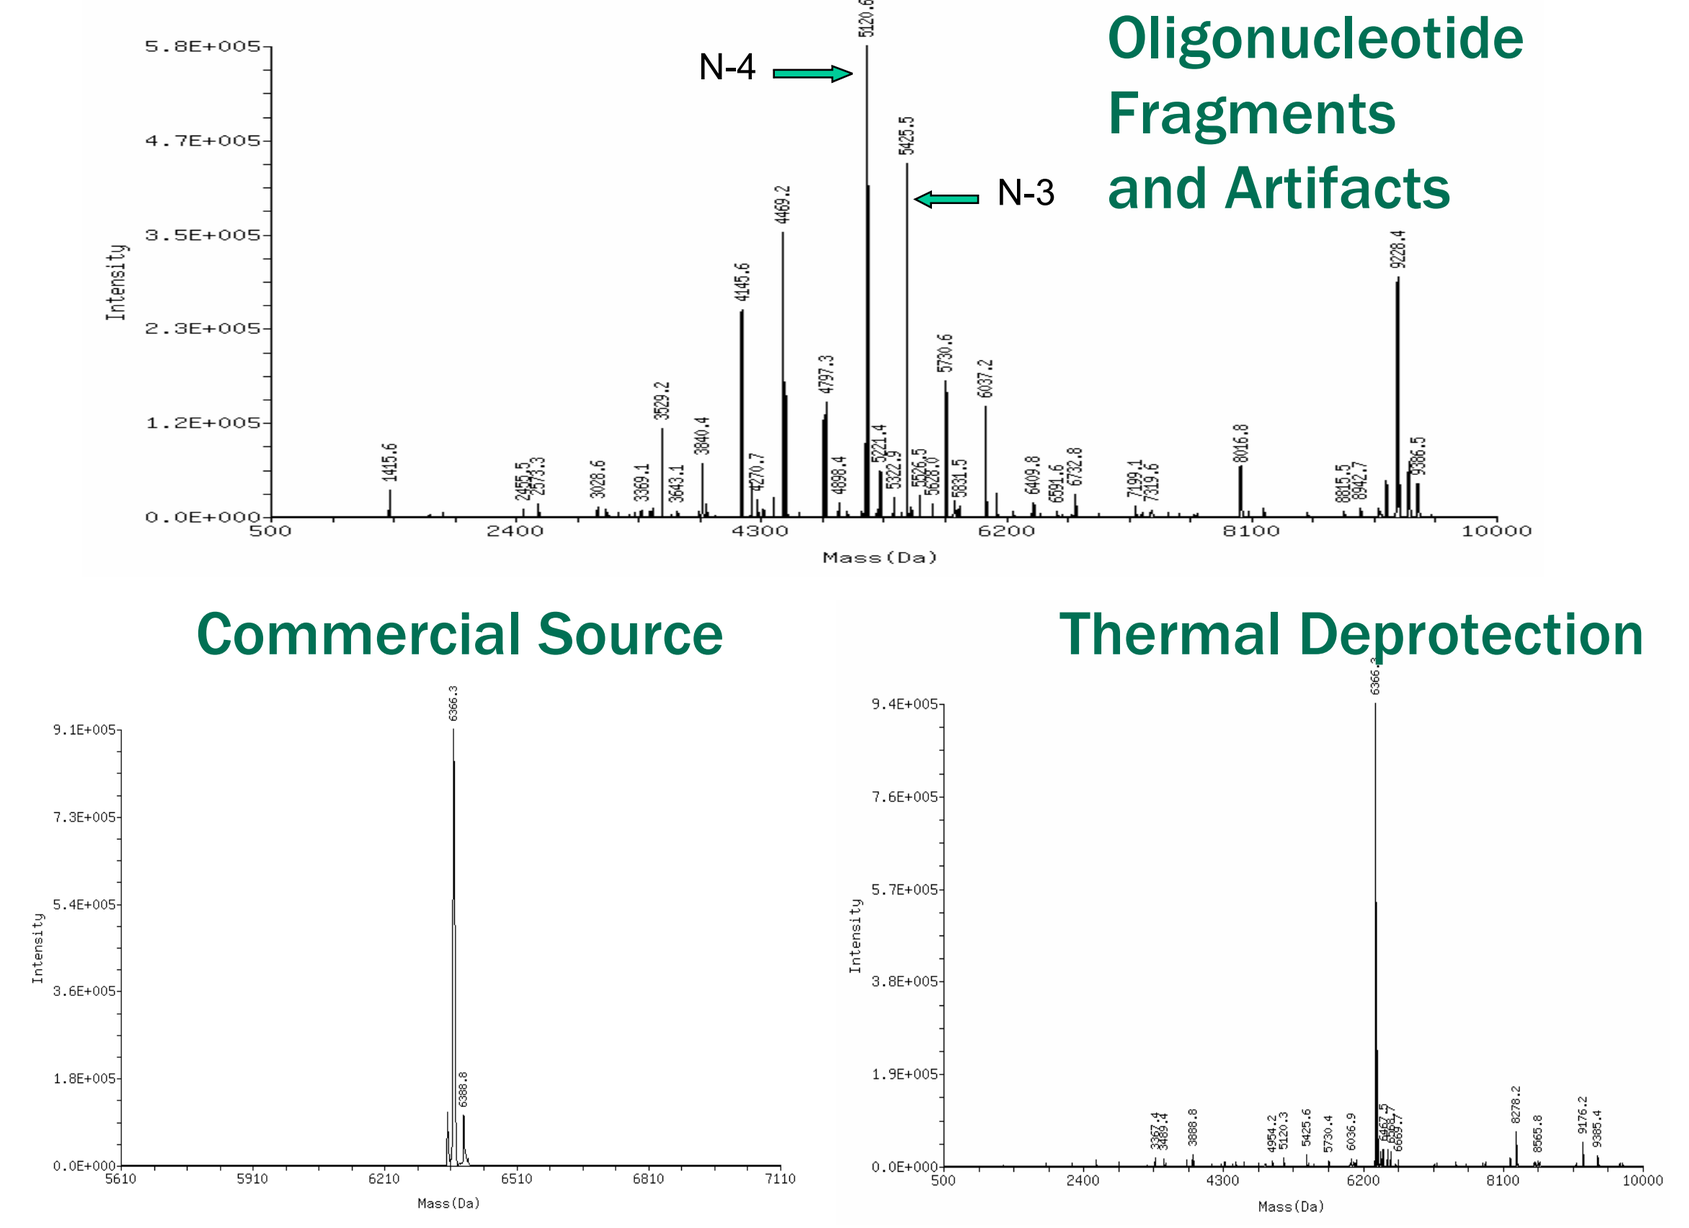

N-4
N-3
Oligonucleotide Fragments and Artifacts
Commercial Source
Thermal Deprotection
Mass (Da)

Deconvoluted MS chromatograms: Range 11.51 to 11.85 min (top); RNA 20 mer from commercial source (below left); RNA 20 mer from Beaucage Synthesis (below right)

Analysis of RNA 20-mer at pH 7.0

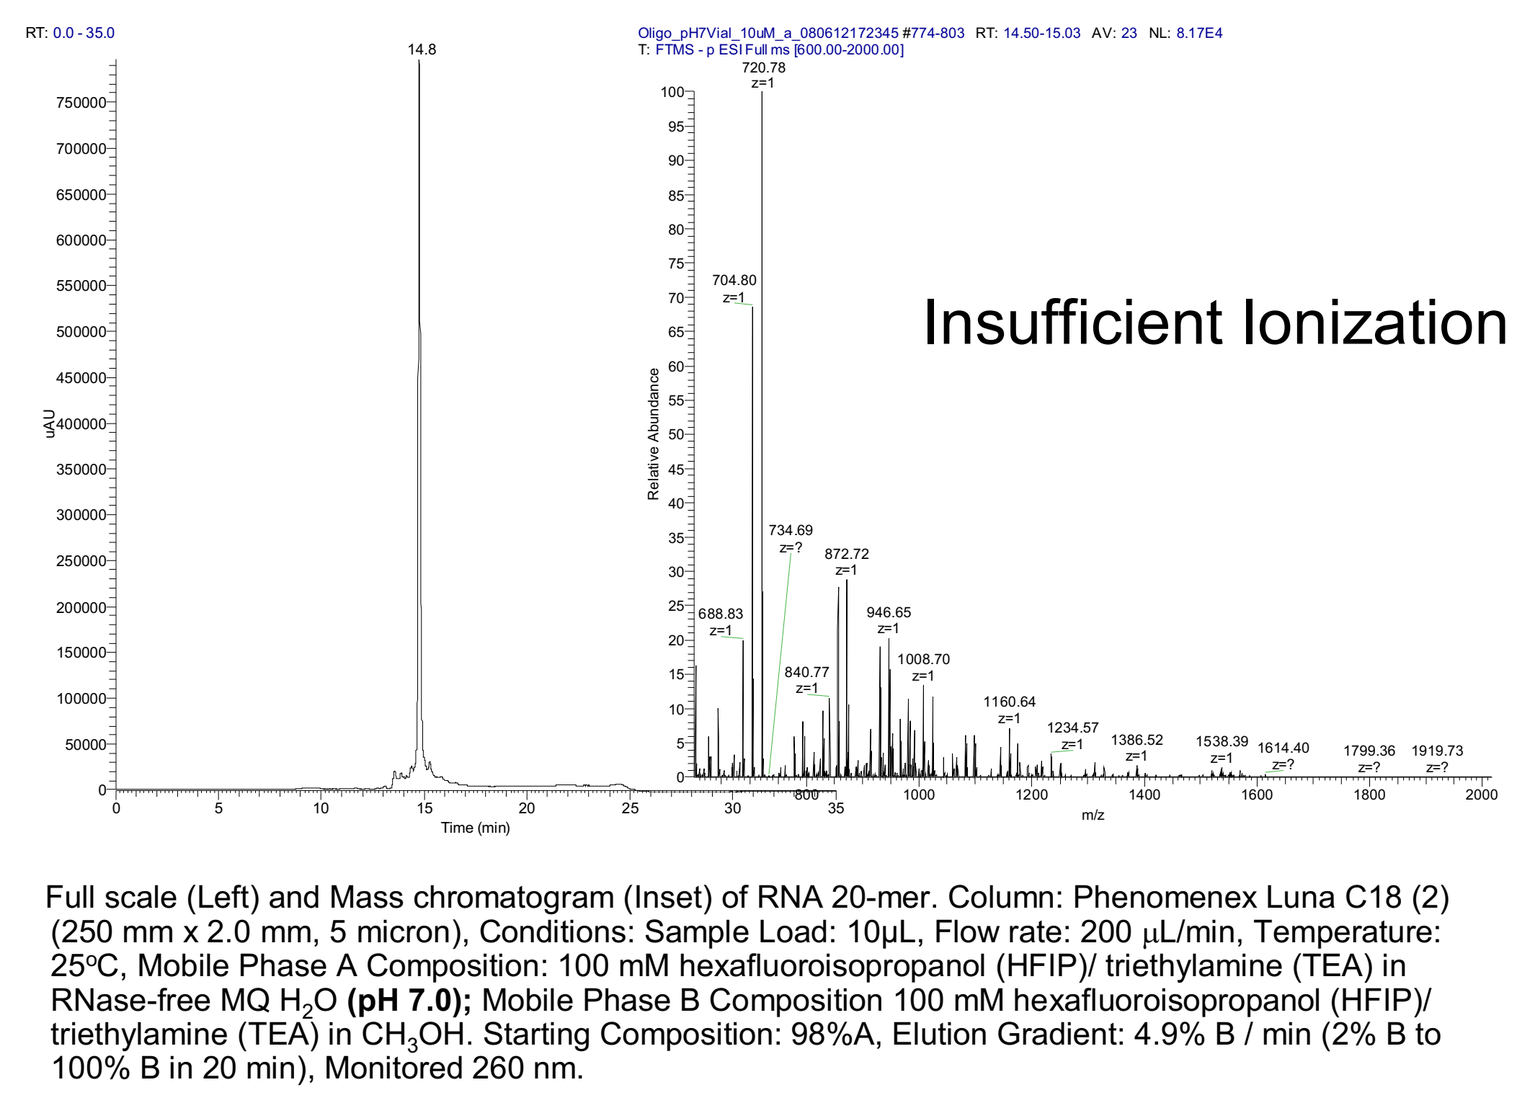

Insufficient Ionization

Full scale (Left) and Mass chromatogram (Inset) of RNA 20-mer. Column: Phenomenex Luna C18 (2) (250 mm x 2.0 mm, 5 micron), Conditions: Sample Load: 10µL, Flow rate: 200 µL/min, Temperature: 25ºC, Mobile Phase A Composition: 100 mM hexafluoroisopropanol (HFIP)/ triethylamine (TEA) in RNase-free MQ H_2O **(pH 7.0);** Mobile Phase B Composition 100 mM hexafluoroisopropanol (HFIP)/ triethylamine (TEA) in CH_3OH. Starting Composition: 98%A, Elution Gradient: 4.9% B / min (2% B to 100% B in 20 min), Monitored 260 nm.

Precision and Limits

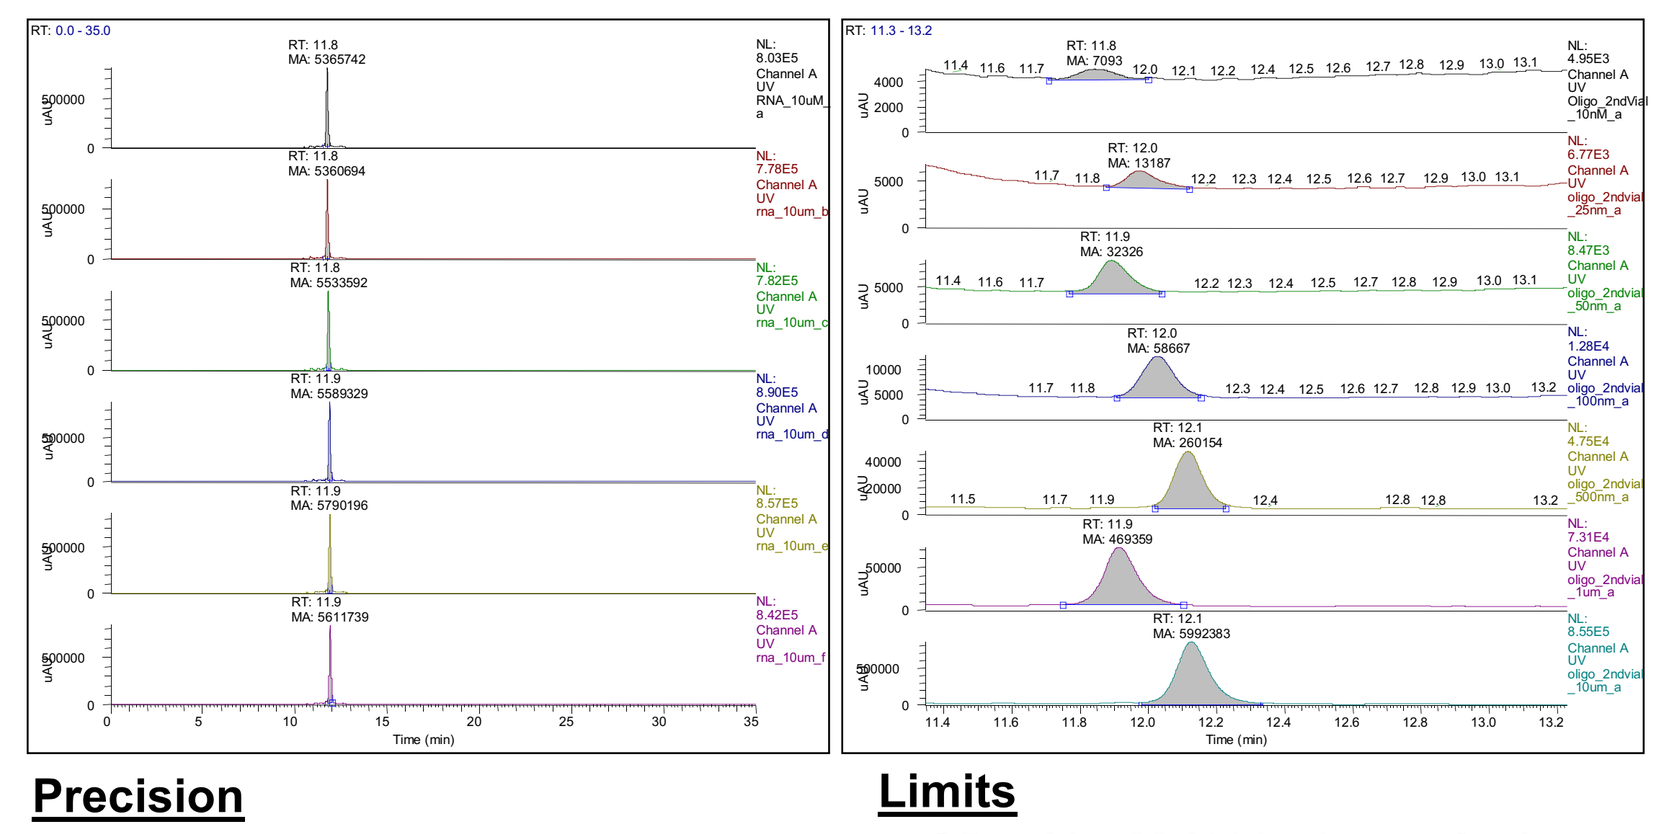

Precision

- **Intra-day 2.94% RSD**
- **Inter-day 3.04% RSD**

Limits

- **LOD ~10 nM (100 picomoles)**
- **LOQ ~25 nM**

(Limits based on signal-to-noise)

MSⁿ: MS/MS of -8 Charge State

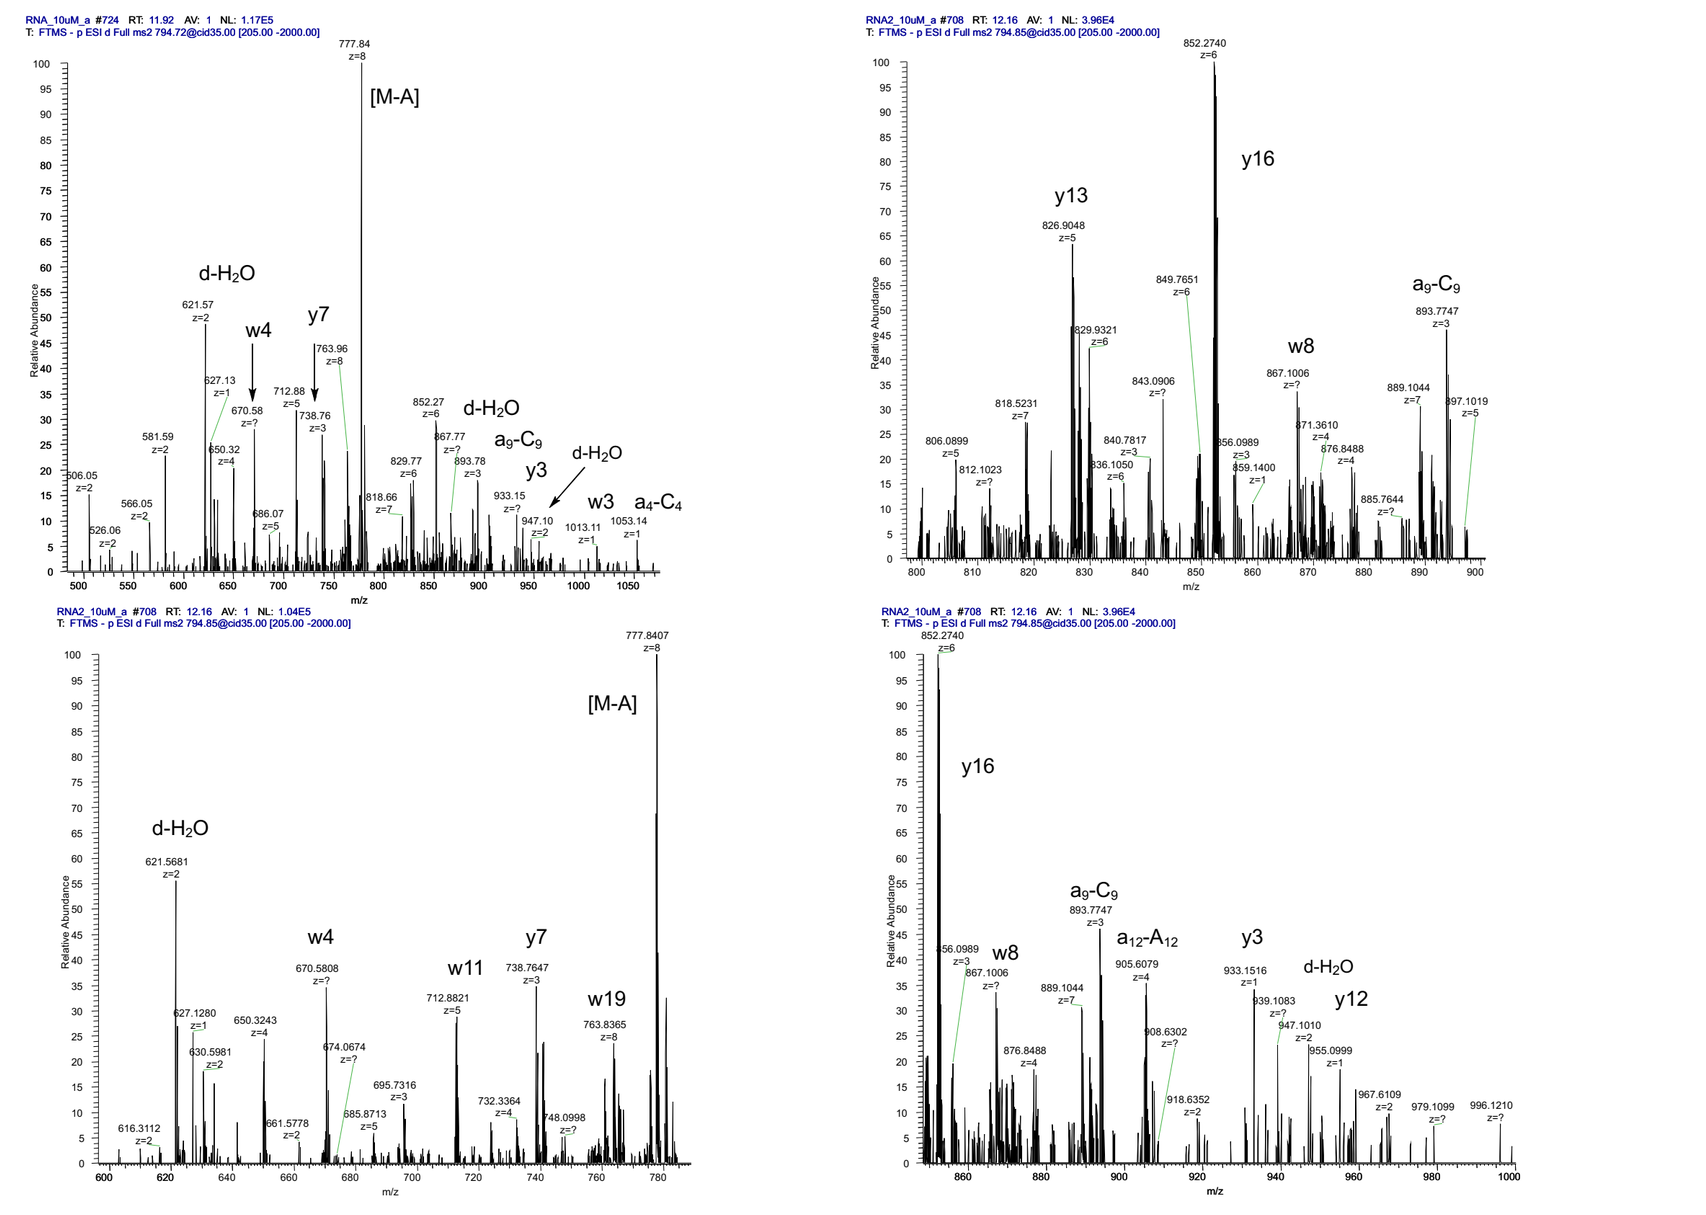

[M-A]
y13
y16
w8

CONCLUSIONS

- The advantage using an LTQ-Orbitrap to confirm the sequence identity of the oligoribonucleotides is its ability to provide high-resolution mass data with the LC-MS analysis.
- Both oligoribonucleotides were comparable with respect to sequence identity. Finally, the method is robust with respect to linearity, accuracy and its ability to separate then identify the failed synthetic sequences (N-1, N-2 etc. oligomers) and product-related impurities. This is the first report of the use of a high resolution hybrid linear ion trap instrument with an oligonucleotide method qualification.

References

- Cieslak, J.; Grajkowski, A.; Kauffman, J.S.; Duff, R.J.; Beaucage, S. L. "The 4-(N-Dichloroacetyl-N-methylamino)benzyloxymethyl Group for 2'-Hydroxyl Protection of Ribonucleotides in the Solid-Phase Synthesis of Oligoribonucleotides" *J. Org. Chem.* **2008**, *73(7)*, 2774-2783.
- Dai, G.; Wei, X.; Liu, Z.; Liu, S.; Marcucci, G.; Chan, K. K. "Characterization and quantification of Bcl-2 antisense G3139 and metabolites in plasma and urine by ion-pair reverse phase HPLC coupled with electrospray ion-trap mass spectrometry" *J. Chromatogr. B.* **2005**, *825*, 201-213.
- Gilar, M.; Fountain, K. J.; Budman, Y.; Holyoke, J. L.; Davoudi, H.; Gebler, J.C. "Characterization of Therapeutic Oligonucleotides Using Liquid Chromatography with On-line Mass Spectrometry Detection" *Oligonucleotides* **2003**, *13*, 229-243.

Acknowledgements

- Serge Beaucage, Ph.D., FDA Center for Drug Evaluation and Research
- Craig P. Dufresne, Ph.D. ThermoFisher
- Brandi L.Conroy, Lancaster Laboratories, Inc